Network Dynamical Systems and Inference for Models of Alzheimer's Disease

Pavanjit Chaggar[1], Gregory Klein[3], Stefano Magon[3], Saad Jbabdi[2], Alain Goriely[1]

1. Mathematical Institute, University of Oxford, 2. WIN, University of Oxford, 3. Hoffmann-La Roche

UNIVERSITY OF OXFORD

Mathematical Institute

Summary

- Alzheimer's disease (AD) is a debilitating neurodegenerative disorder whose pathology remains poorly understood.
- Toxic tau-protein is a key driver of AD pathology and has a well characterised staging profile (shown below) that is highly correlated with brain atrophy and symptom onset.
- Instead of using expensive PDE systems, we use a network based model to describe AD.
- The reduced cost of the network model relative to its PDE counterpart allows us to use MCMC inference to calibrate personalised patient predictions.

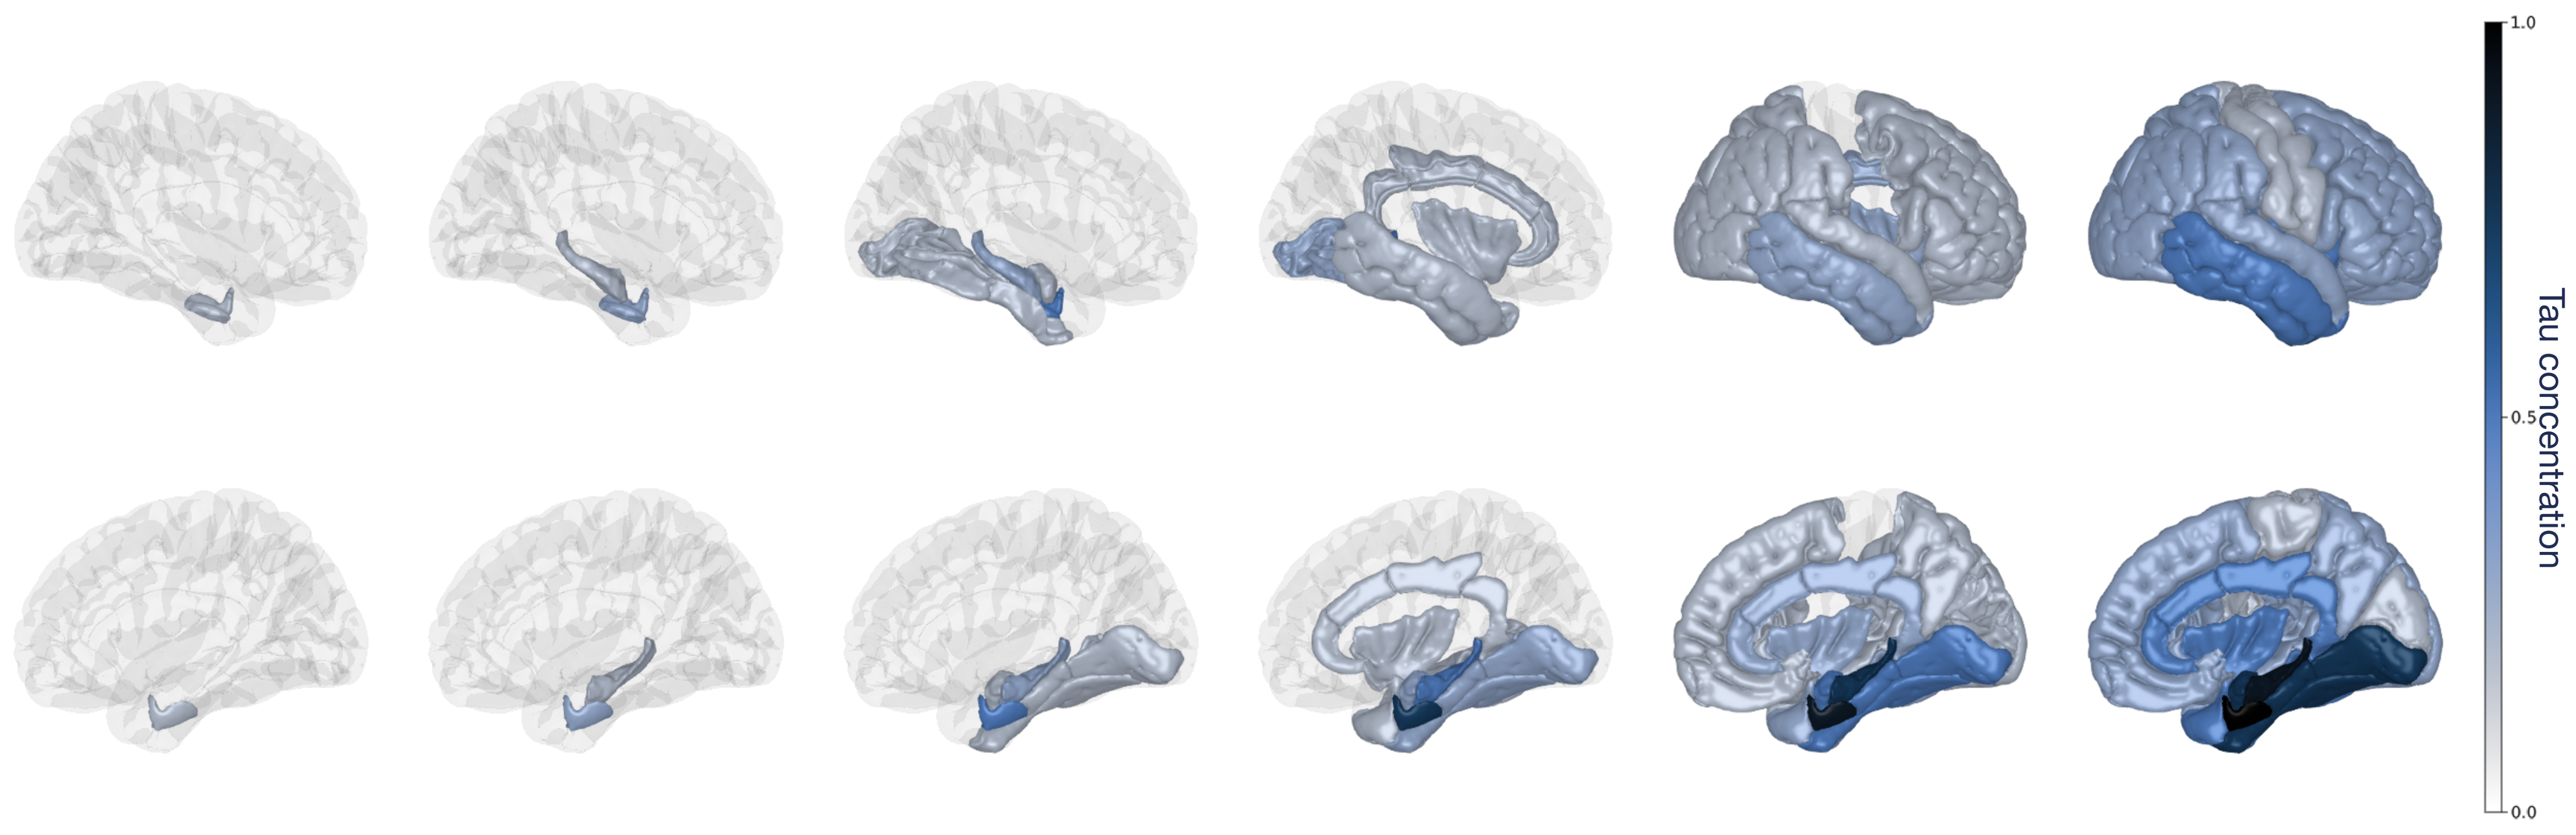

1.0
0.5
0.0
Tau concentration

Modelling AD on a Network

We model toxic protein dynamics in AD with two processes: transport through the brain and a local autocatalytic growth process.

- We model transport as diffusion on a network using a graph Laplacian, $\mathbf{L} = \mathbf{D} - \mathbf{A}$, with degree matrix, $\mathbf{D}$, and adjacency matrix, $\mathbf{A}$, derived from diffusion tractography and representing axonal connections between regions.

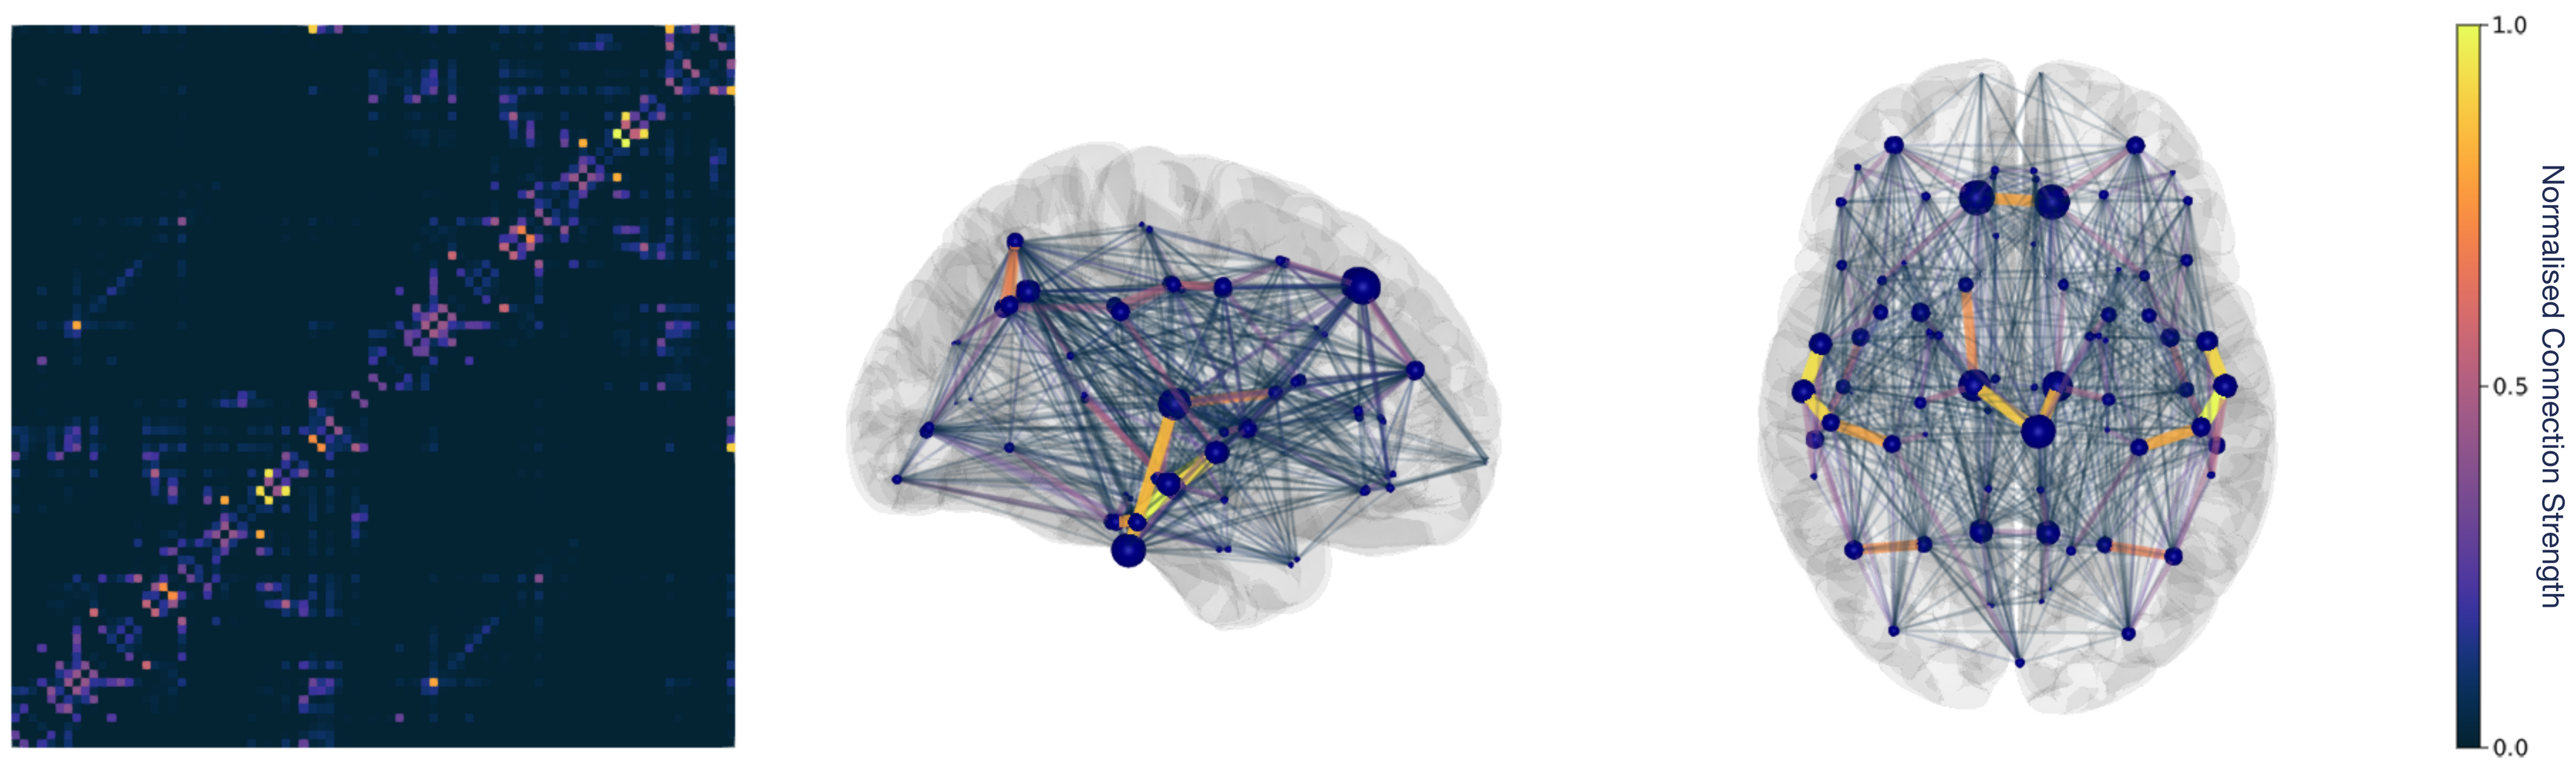

1.0
0.5
0.0
Normalised Connection Strength

- To model growth, we use a quadratic growth term regionally bounded by a healthy base concentration, $\mathbf{p_0}$, and a carrying capacity, $\mathbf{p}_\infty$.

$$\frac{dp_i}{dt} = \underbrace{-\rho L_{ij}(p_j - p_{0,j})}_{transport} + \underbrace{\alpha(p_i - p_{0,i})\left(1 - \frac{p_i}{p_{\infty,i}}\right)}_{growth}$$

We use a gaussian mixture model on population data to separate out healthy and pathological tau-PET signal.

- $\mathbf{p_0}$ is taken to be the mean of the inferred healthy distribution.
- $\mathbf{p}_\infty$ is approximated as the 99th quantile of the inferred pathological distribution.

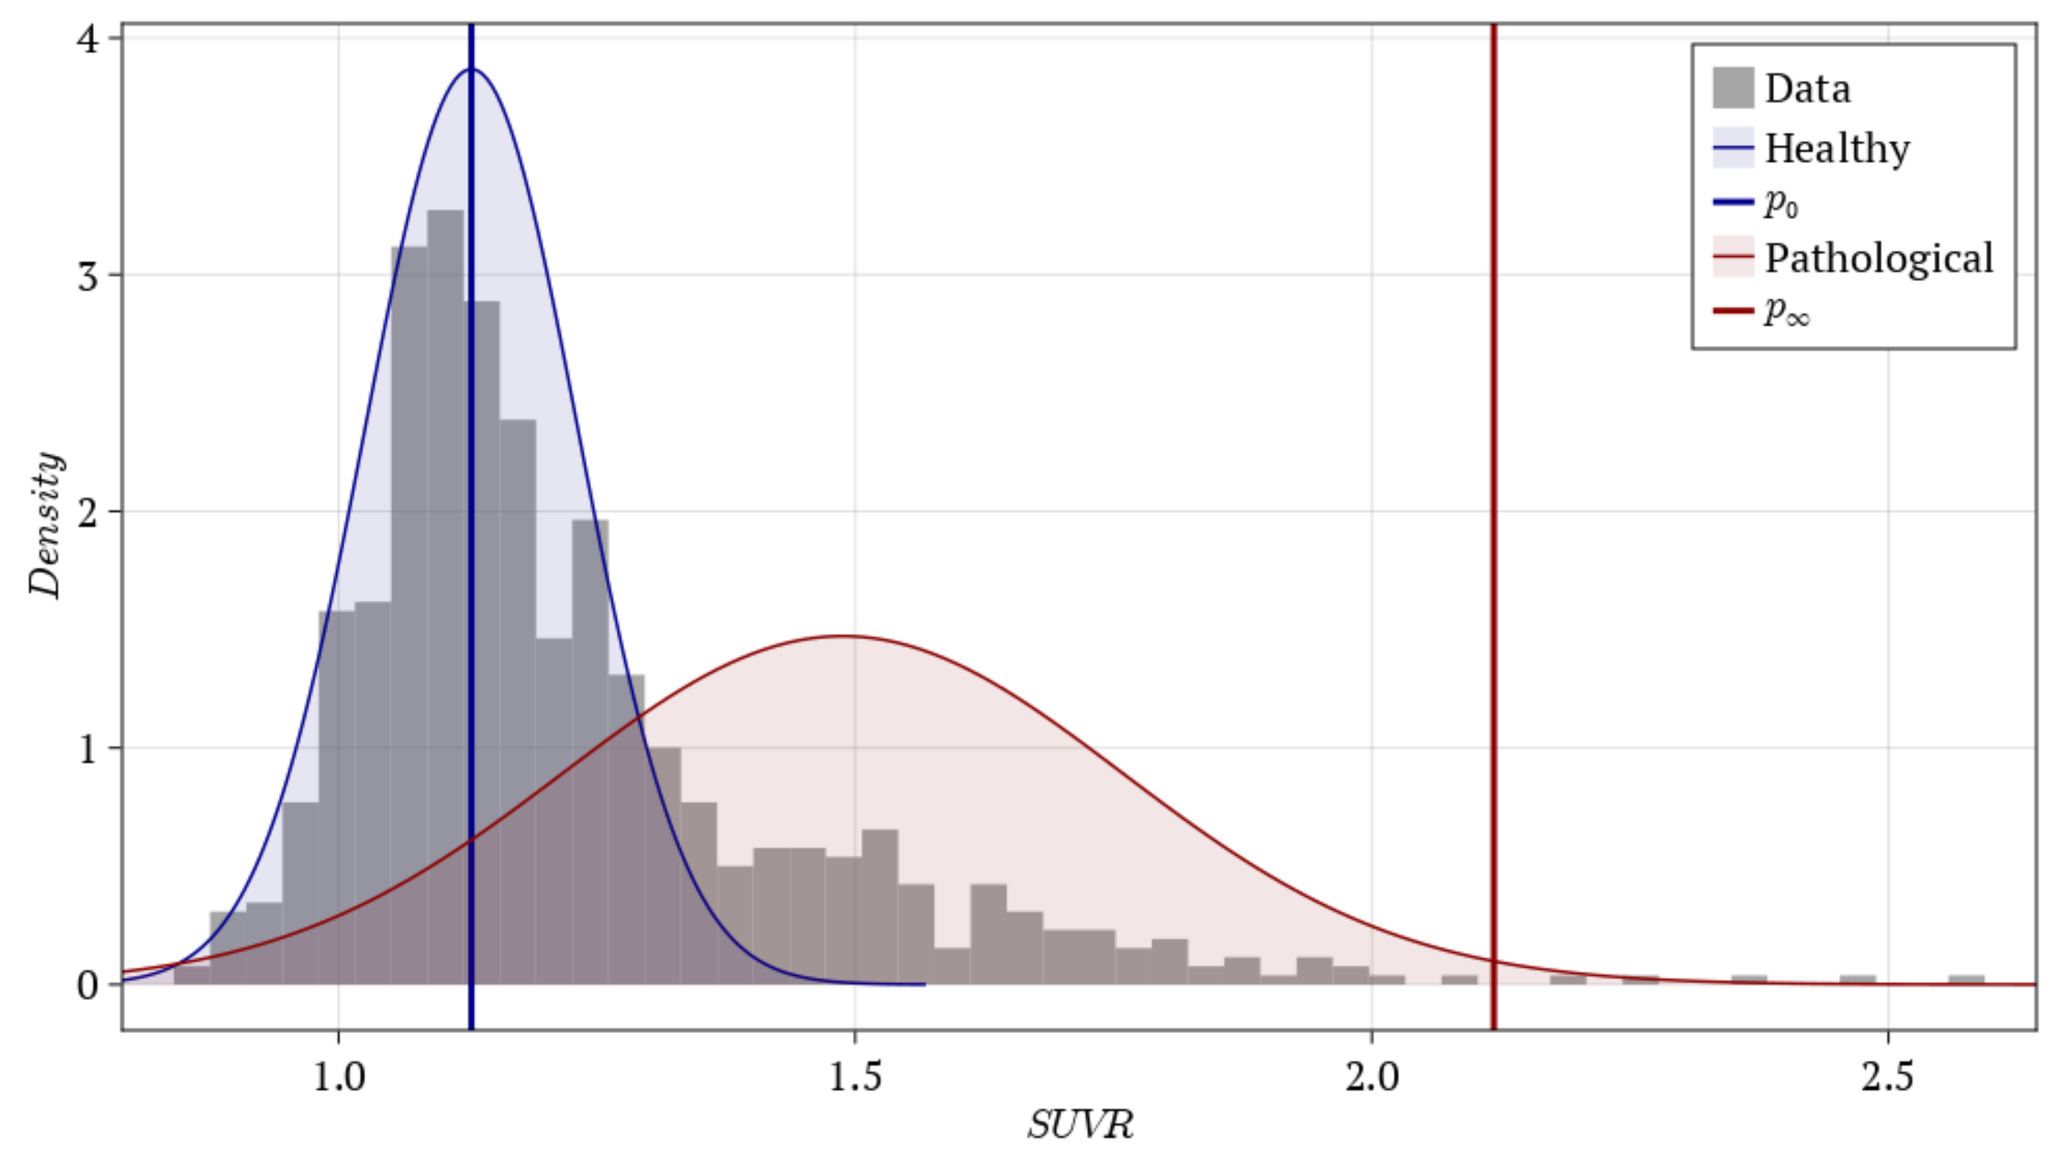

Data
Healthy
p_0
Pathological
p_∞
Density
SUVR

Probabilistic Models and Inference

Here we present a case study of an AD positive subject from the Alzheimers Disease Neuroimaging Initiative (ADNI) dataset.
We use pre-analysed data from ADNI which are summarised across 83 regions for each of 4 scans over a period of 3.5 years. The data, $\hat{\mathbf{y}}$, is a 83×4 dimensional, for 83 parcellated brain regions and 4 scans.

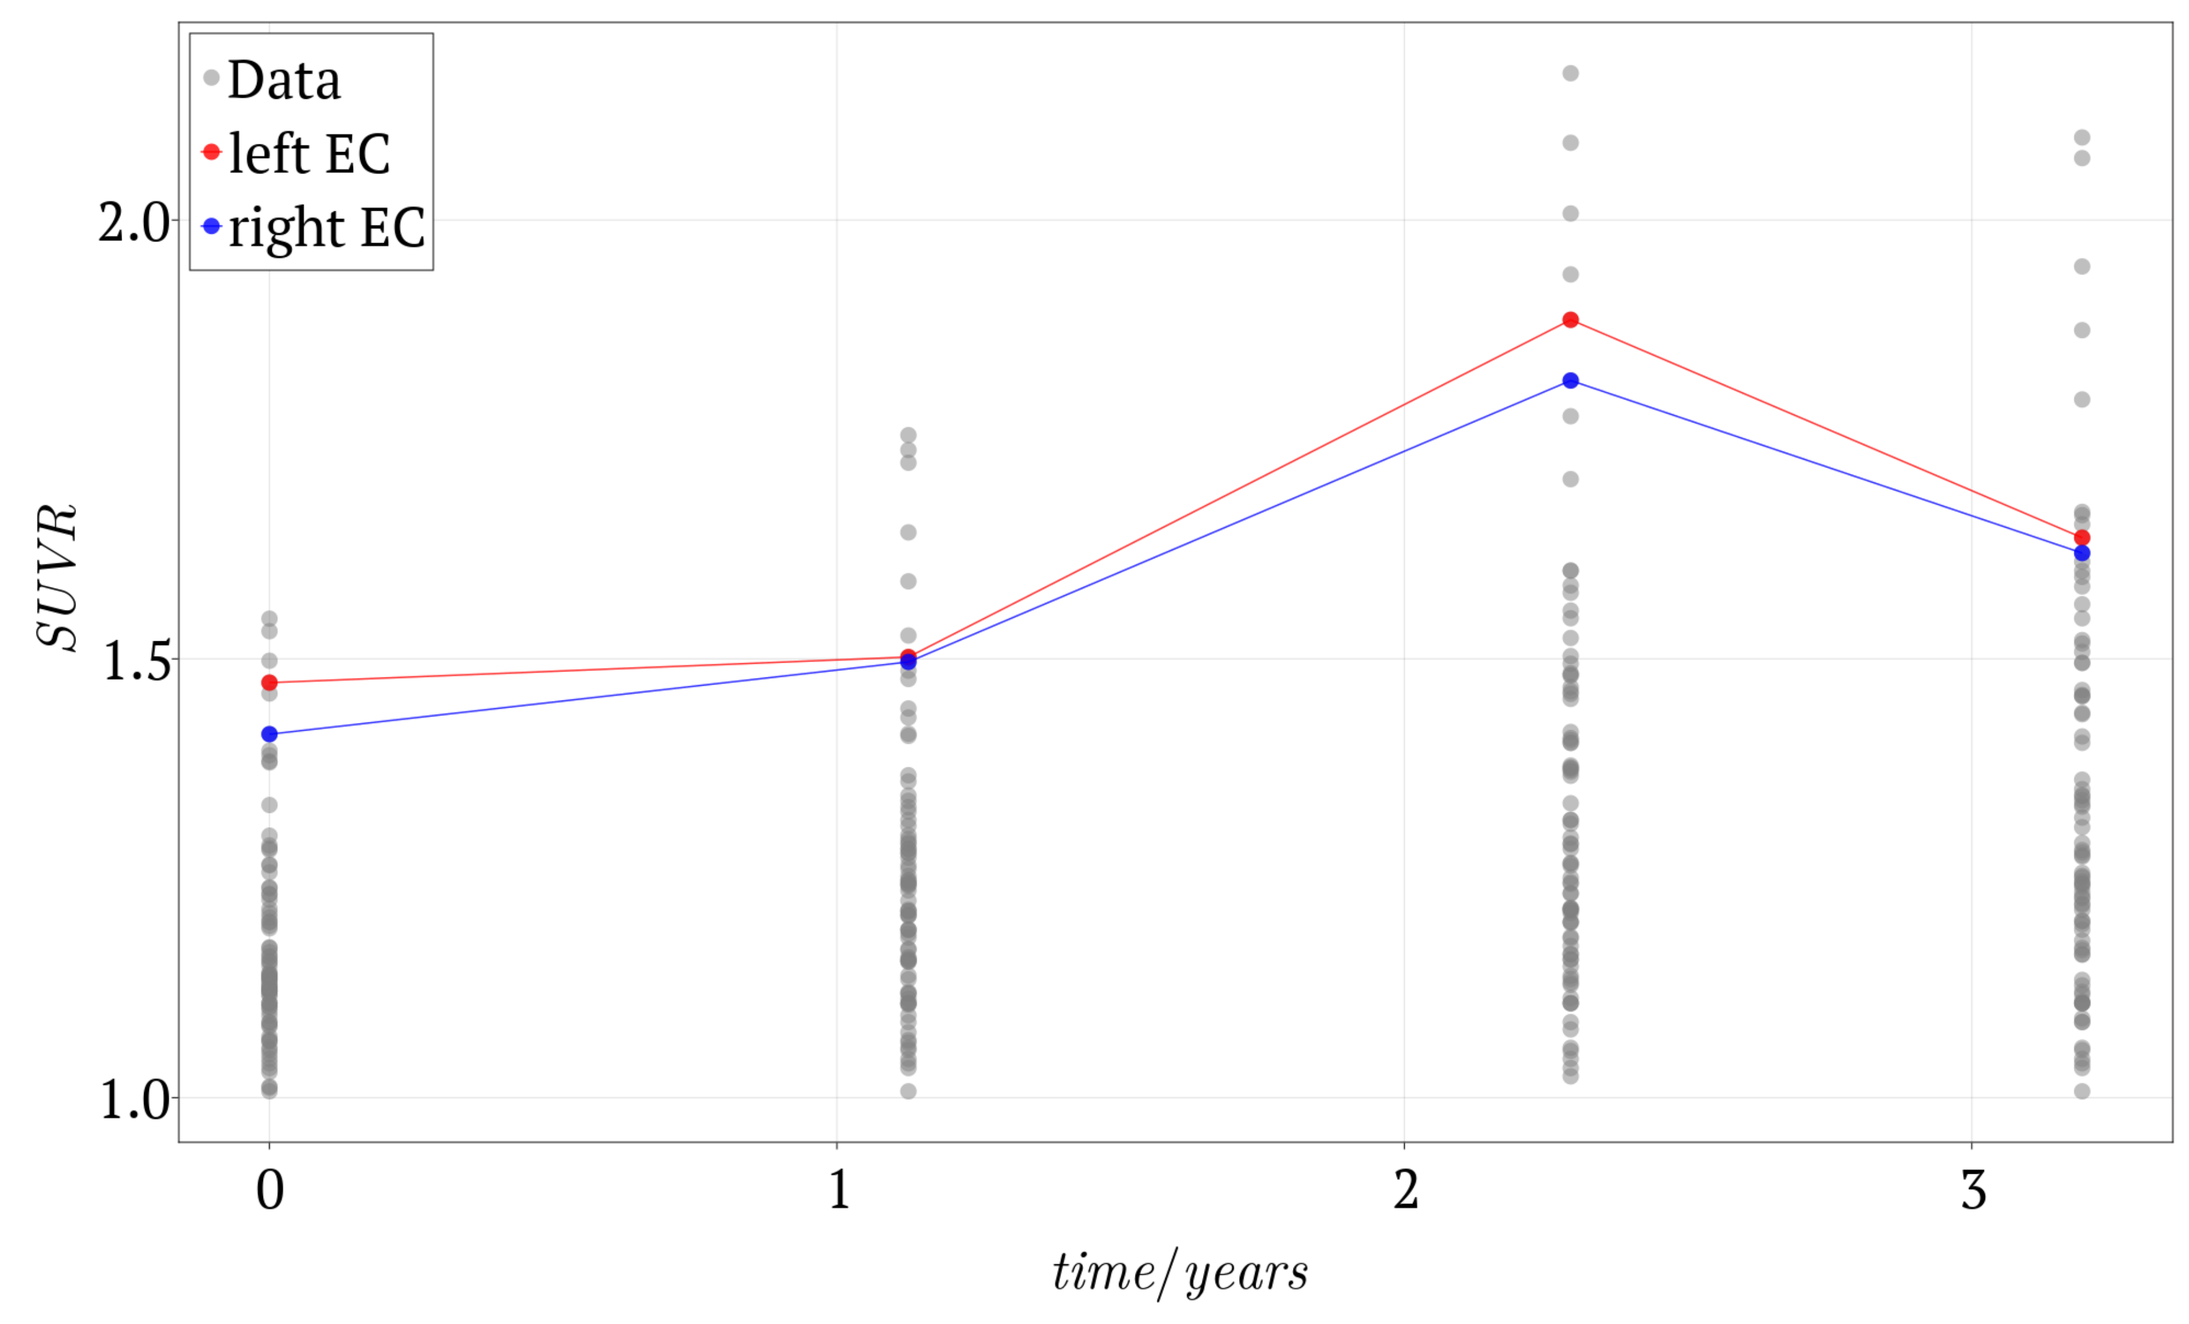

Data
left EC
right EC
SUVR
time/years

We assume the following data-generating model with Gaussian i.i.d noise.

$$g(\mathbf{y_0}, \theta, t, \sigma) = f(\mathbf{y_0}, \theta, t) + \mathcal{N}(0,\sigma)$$

where $\mathbf{y}_0 \in \mathbb{R}^N$ are the initial conditions for N nodes, $\theta \in \mathbb{R}^2$ are the model parameters, $\mathbf{t} \in \mathbb{R}^T$ is observation times for T scans, σ is the standard deviation of the measurement noise. We wish to estimate the posterior over $\mathbf{y}_0, \theta$ and σ:

$$p(\mathbf{y}_0, \theta, \sigma \mid \hat{\mathbf{y}}, t) \propto \prod_j^T \prod_i^N p(\hat{y}_{ij} \mid \mathbf{y}_0, \theta, \sigma, t_j) p(\mathbf{y_0}, \theta, \sigma)\,.$$

We use the following priors:

$$\begin{aligned} \mathbf{y}_0 &\sim \mathcal{N}(\hat{\mathbf{y}}_0, 0.5, [\mathbf{p}_0, \mathbf{p}_\infty]) \\ \rho &\sim \mathcal{N}(0,5,[0,\infty]) \\ \alpha &\sim \mathcal{N}(0,5) \\ \sigma &\sim \Gamma^{-1}(2,3) \end{aligned}$$

To perform inference, we use a No-U-Turn Sampler with a target acceptance rate of 0.8 and collected 3 chains of 3000 samples.

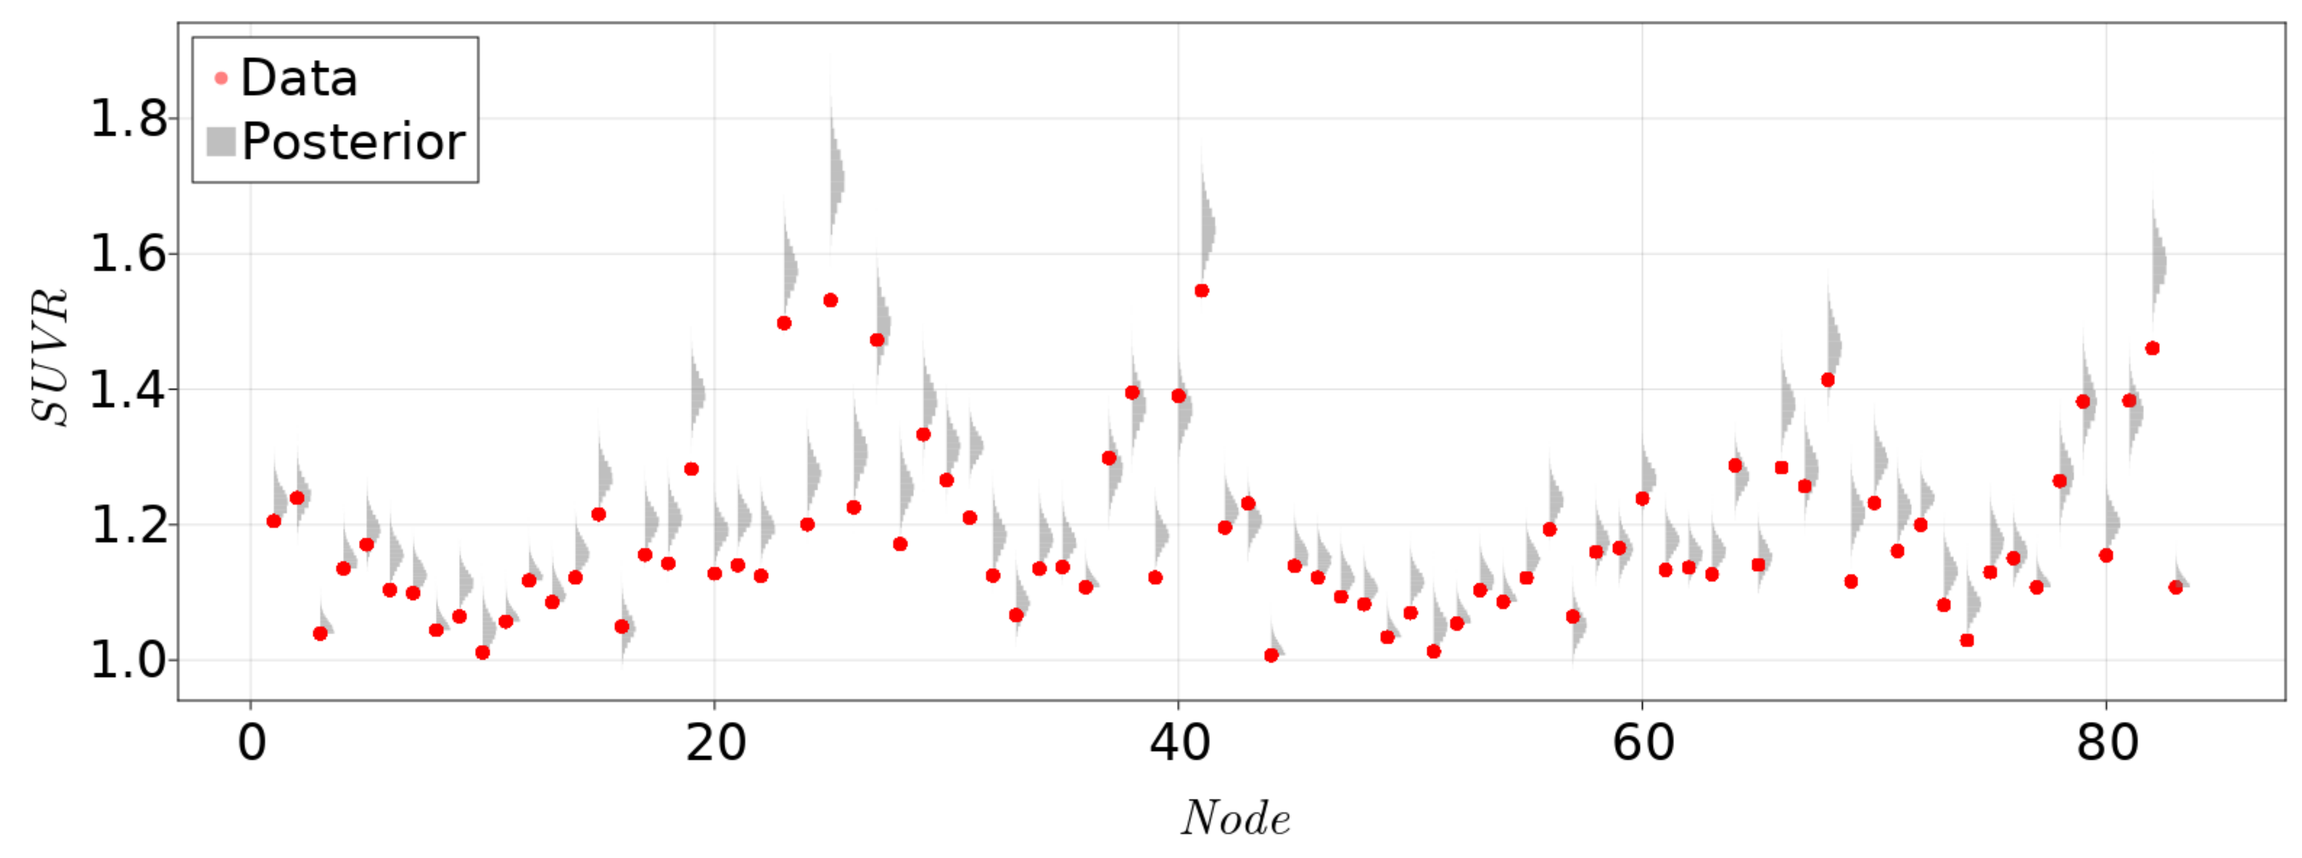

Data
Posterior
SUVR
Node

- For $\mathbf{y}_0$, the posteriors are tightly aligned with the corresponding data point, with notable exceptions for subcortical regions, where there is known to be substantial off target binding.
- For model parameters, ρ and α, the posteriors show that the system is primarily driven by growth, as opposed to diffusion. This is consistent with previous results using hierarchical inference to show that for population parameters growth dominates over diffusion.
- Additionally, we see that there is substantial noise in the system. This could simply be because PET is noisy, or because the system is not expressive enough, e.g. cannot capture non-monotonic trajectories.

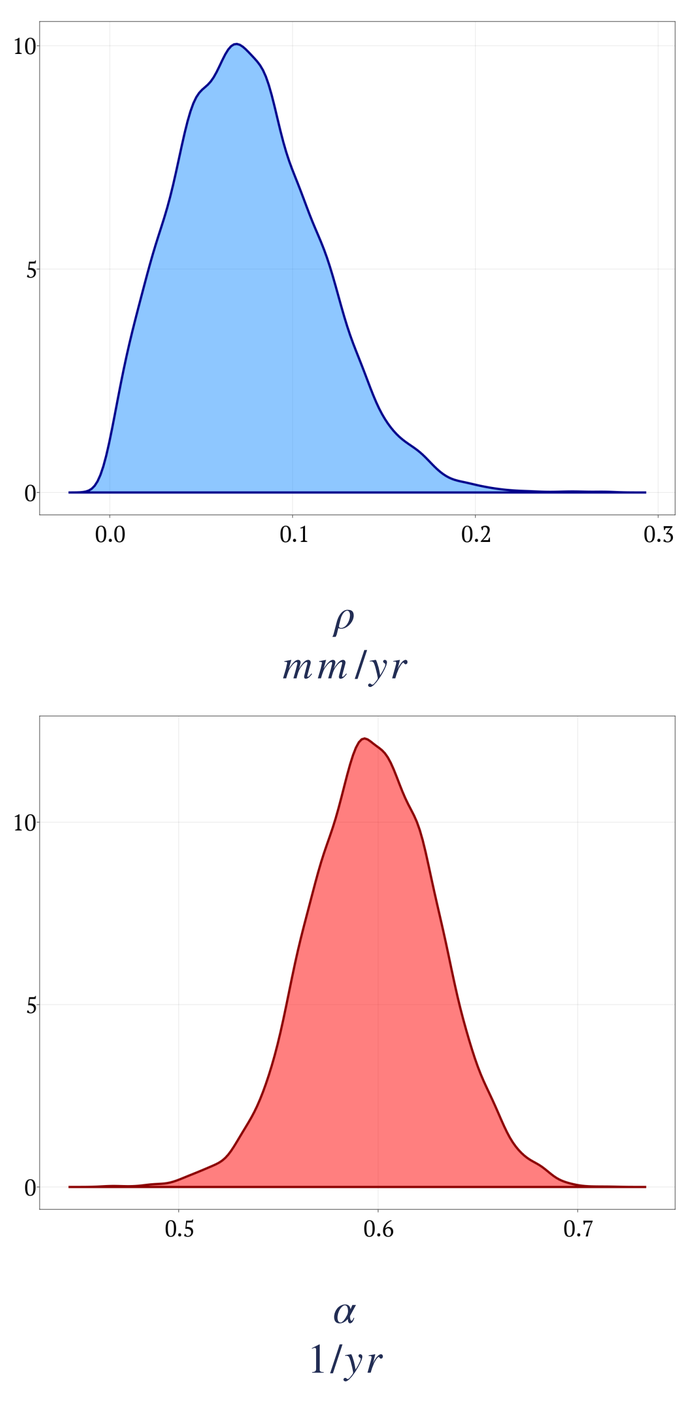

ρ
mm/yr
α
$1/yr$

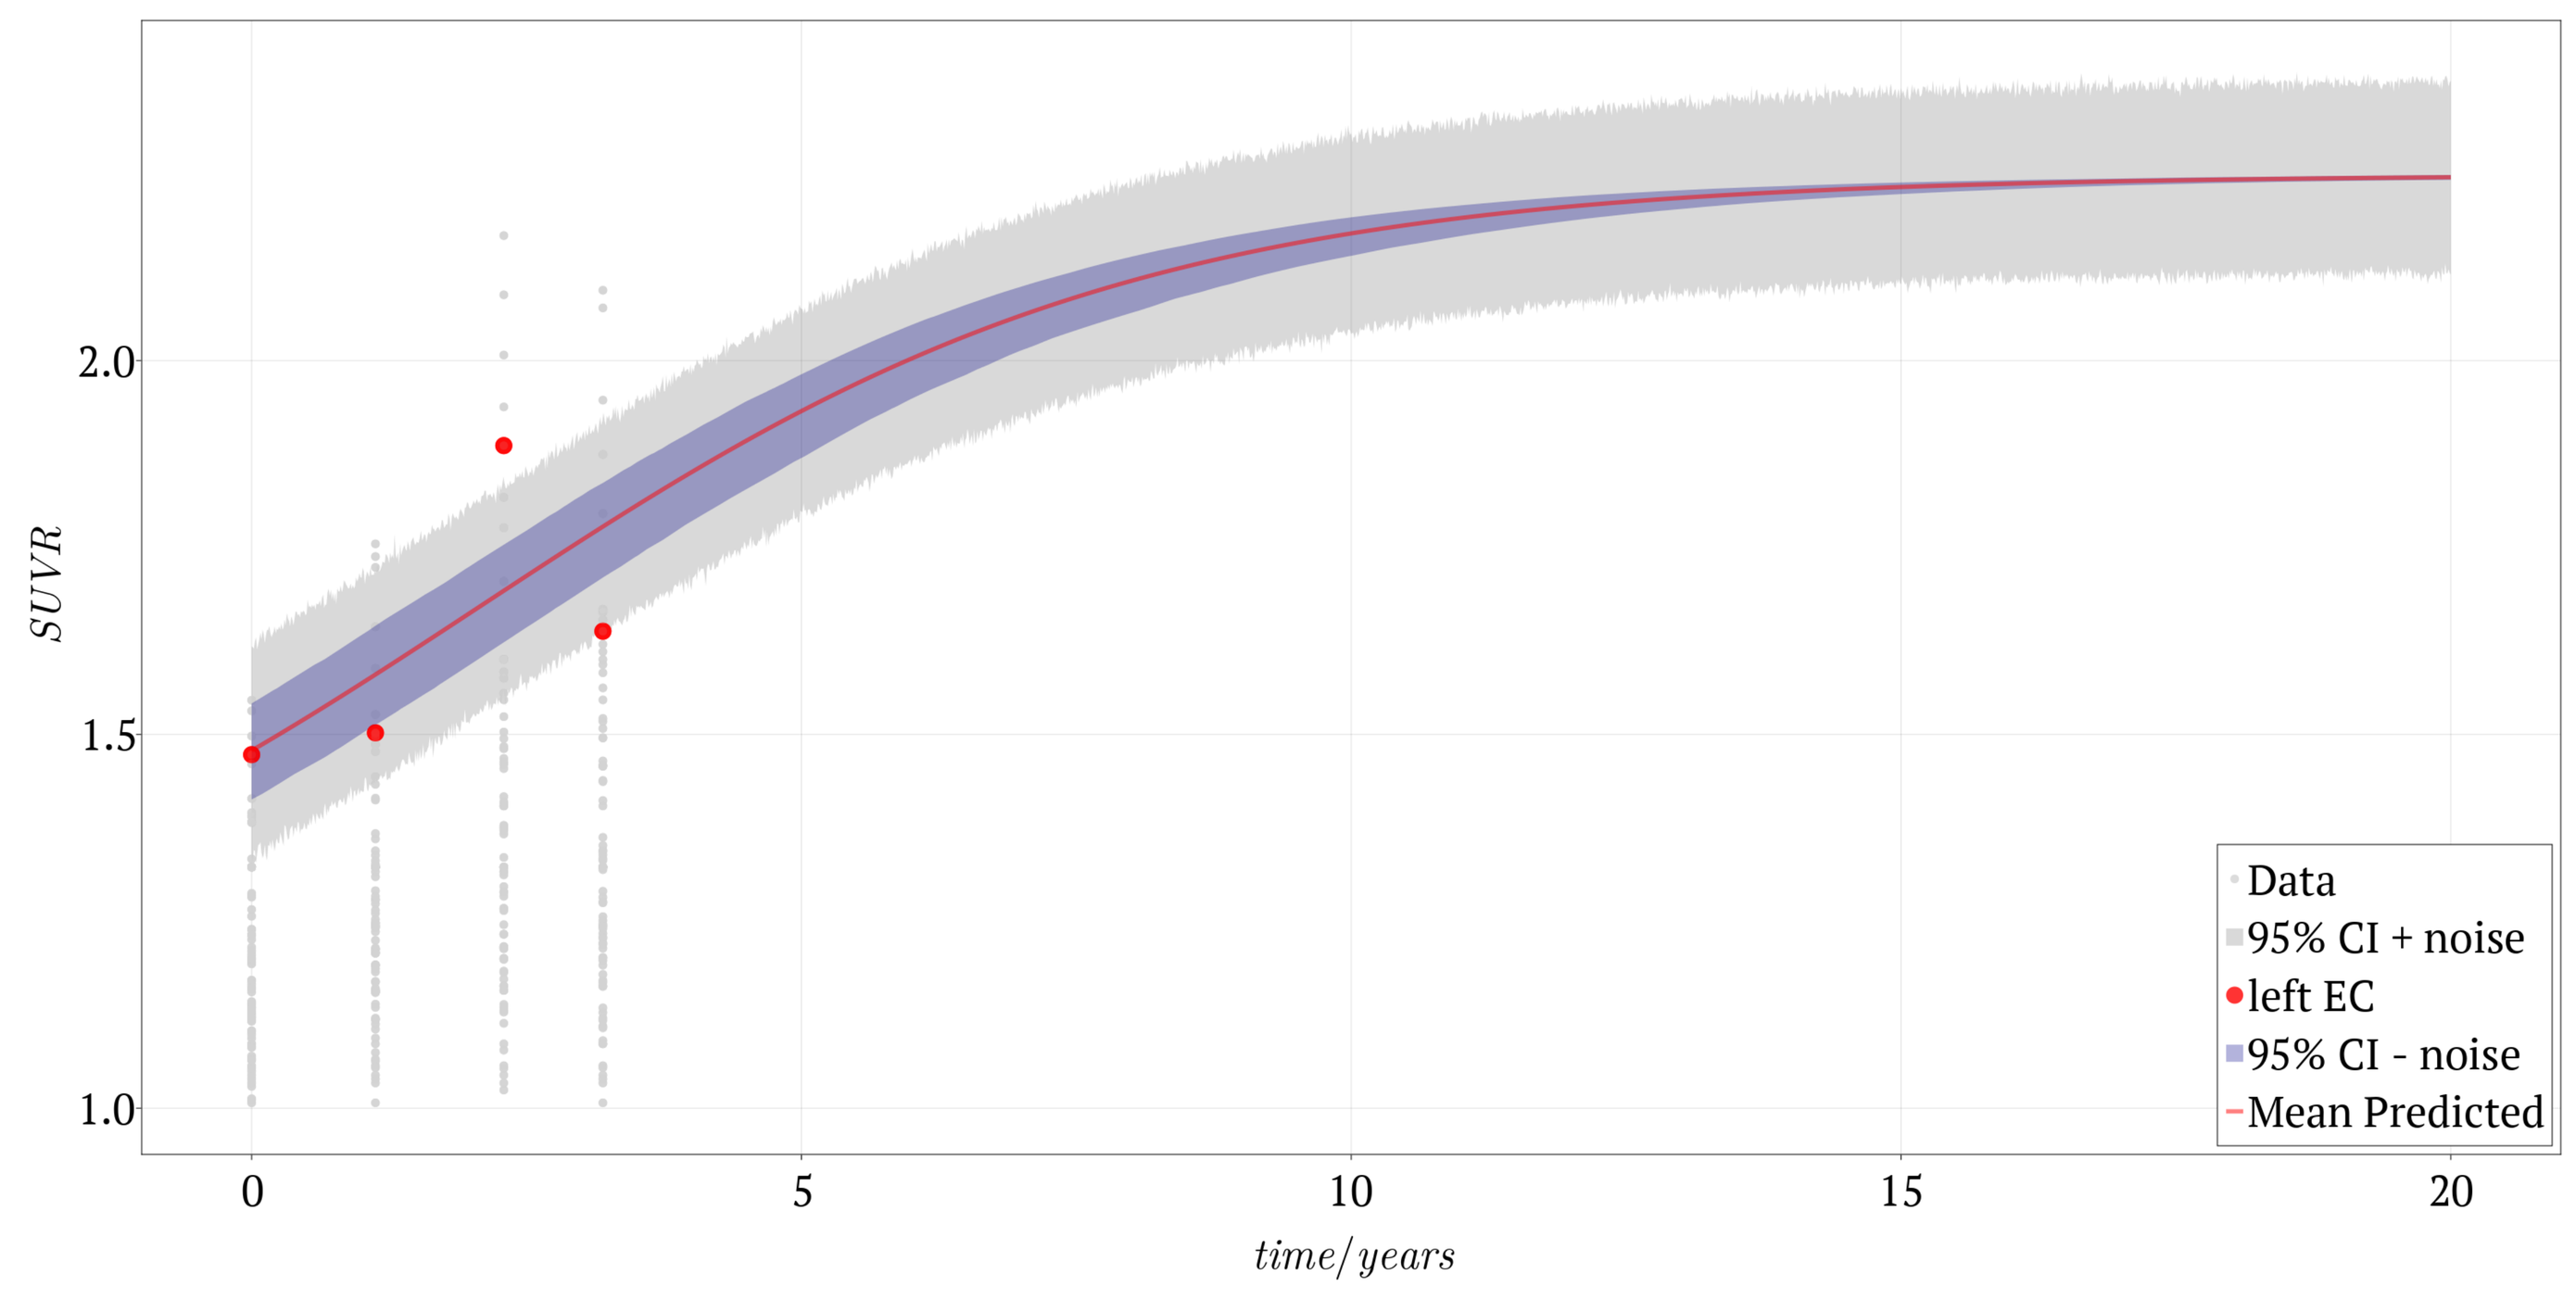

Data
95% CI + noise
left EC
95% CI - noise
Mean Predicted
SUVR
time/years

References:
Prion-like spreading of Alzheimer's disease within the brain's connectome S Fornari, A Schäfer, M Jucker, **A Goriely,** E Kuhl, Journal of the Royal Society Interface, 2019
Correlating tau pathology to brain atrophyusing a physics-based Bayesian model, A Schafer, **P Chaggar, A Goriely**, E Kuhl, Engineering with Computers, 2022 (Joint first author).

Oxford Mathematics